Assessment of AMX0114, An Antisense Oligonucleotide Targeting Calpain-2, in Multiple Models of Axonal Degeneration

Evan Mizerak,[1] Joshua Cohen,[1] Justin Klee,[1] Michael Bekier,[2] Sami Barmada,[2] Kevin McAvoy,[3] Hibiki Kawamata,[3]

[1]Amylyx Pharmaceuticals, Inc., Cambridge, Massachusetts; [2]Michigan Medicine, Ann Arbor, Michigan; [3]Weill Cornell Medicine, New York, New York

BACKGROUND

- Axonal degeneration is a key contributor to the clinical presentation and pathogenesis of amyotrophic lateral sclerosis (ALS) and other neurodegenerative diseases[1,2]
- Activation of the calcium-dependent protease calpain-2 is a critical effector of axonal degeneration and neuronal cell death (**Figure 1**)[2,3]
- Calpain-2 is implicated in the pathogenesis of ALS based on:
 - Findings of elevated calpain-2 messenger RNA (mRNA) in muscle samples[4] and calpain-specific transactive response DNA-binding protein 43 (TDP-43) cleavage product concentrations in postmortem spinal cord[3,5] and brain[3] samples from people with ALS
 - Calpain-dependent TDP-43 cleavage promotes aggregation of TDP-43, a pathologic hallmark in ALS and other neurodegenerative diseases[3]
 - Therapeutic benefit of calpain-2 activity modulation in animal models of ALS[6]
 - The role of calpain-2 in cleaving neurofilament, a component of the axonal cytoskeleton[2] and a broadly researched biomarker in ALS
- Activation of calpain-2 has been implicated in neuronal death resulting from acute neuronal injury[7]
- Amylyx developed AMX0114, an antisense oligonucleotide targeting calpain-2. AMX0114 demonstrates potent, dose-dependent, and durable knockdown of *CAPN2* (gene encoding calpain-2) mRNA and calpain-2 protein, as well as functional efficacy in preclinical models of neurodegeneration

What Are Calpains?

- Calpains are a family of calcium-dependent cysteine proteases that target multiple substrates within the axonal cytoskeleton[2]
- There are >12 calpain isoforms. Of the 2 main isoforms (calpain-1 and calpain-2), calpain-1 is generally believed to play a neuroprotective role, while activation of calpain-2 is associated with axonal degeneration[3,8]

Figure 1. CONSEQUENCES OF CALPAIN ACTIVATION FOR MOTOR NEURON FUNCTION[3]

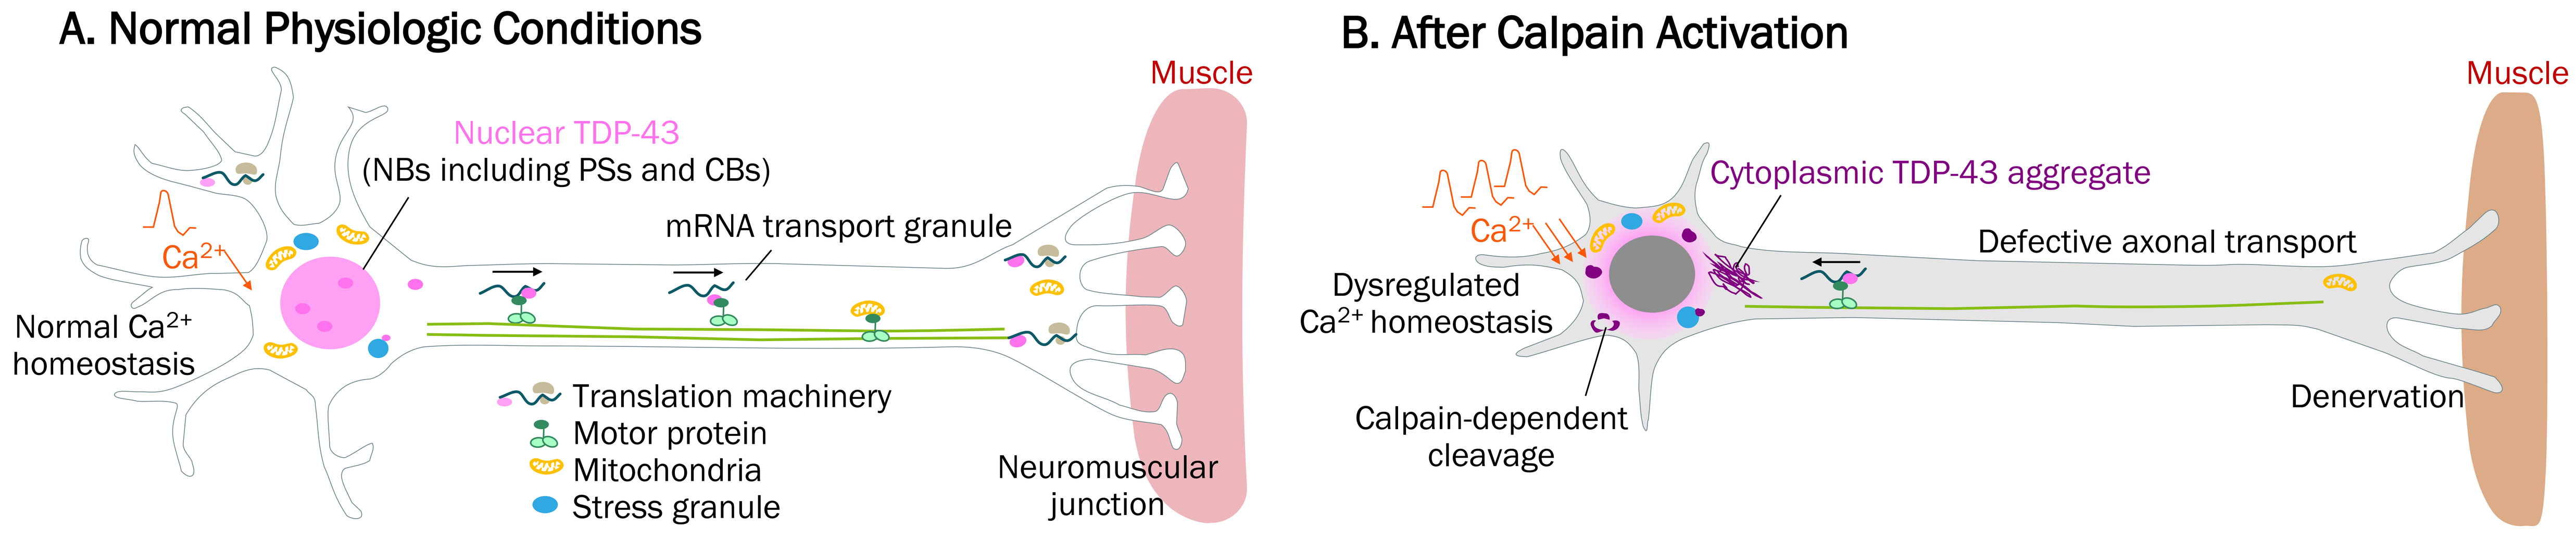
A. Normal Physiologic Conditions
Muscle
Nuclear TDP-43
(NBs including PSs and CBs)
mRNA transport granule
Ca^{2+}
Normal Ca^{2+} homeostasis
Translation machinery
Motor protein
Mitochondria
Stress granule
Neuromuscular junction
B. After Calpain Activation
Muscle
Cytoplasmic TDP-43 aggregate
Ca^{2+}
Defective axonal transport
Dysregulated Ca^{2+} homeostasis
Calpain-dependent cleavage
Denervation

Ca2+, calcium; CB, Cajal body; mRNA, messenger ribonucleic acid; NB, nuclear body; PS, paraspeckle; TDP-43, transactive response DNA-binding protein 43.

OBJECTIVE

- To assess the neuroprotective effects of AMX0114 by evaluating in multiple models of axonal degeneration, including a human neuron model of ALS and a model of acute neuronal injury

EXPERIMENTS

In Vitro Efficacy Assessment: Survival Analysis

METHOD: Assessment of survival at multiple AMX0114 dose levels in a model of mutant TDP-43 pathology

- Human induced pluripotent stem cell (iPSC)-derived neurons (iNeurons) harboring a pathogenic TARDBP mutation (M337V) were treated with AMX0114 at multiple concentrations
- Cumulative risk of death over 10 days was assessed using automated fluorescence microscopy and genetically encoded death indicators (GEDI)
- Levels of extracellular neurofilament light chain (NfL) were assessed by ELISA

- Robust knockdown achieved by AMX0114 translated to dose-dependent improvements in survival in an *in vitro* neuronal ALS model (**Figure 2**)
- The hazard ratio for M337V cells treated with 0.1 μM AMX0114 was 67.8% lower than that of vehicle (DMSO)-treated M337V cells ($p<0.0001$)
- Treatment with 0.1 μM AMX0114 resulted in ~60% decrease in extracellular NfL above baseline relative to DMSO (vehicle)-treated M337V controls **(Figure 3)**
- Decreases in NfL levels from baseline 11 days following treatment with AMX0114 were correlated with decreased risk of death

Figure 2. Dose-Dependent Effect of AMX0114 on Cumulative Risk of Death

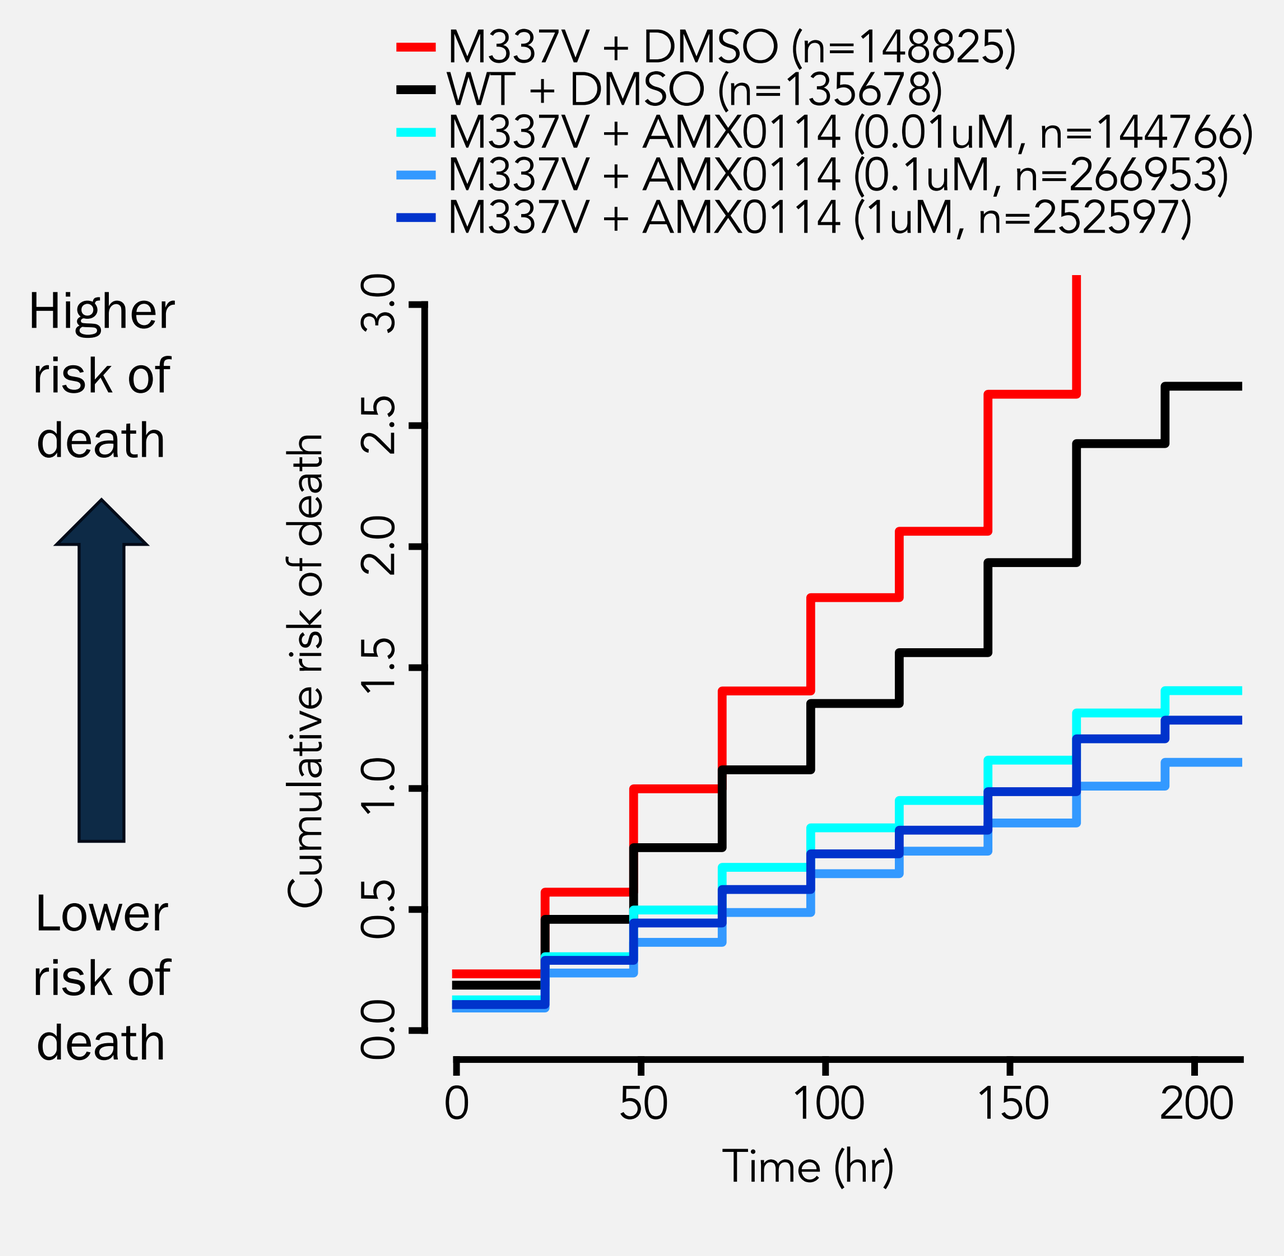
M337V + DMSO (n=148825)
WT + DMSO (n=135678)
M337V + AMX0114 (0.01uM, n=144766)
M337V + AMX0114 (0.1uM, n=266953)
M337V + AMX0114 (1uM, n=252597)
Higher risk of death
Lower risk of death
Cumulative risk of death
0.0
0.5
1.0
1.5
2.0
2.5
3.0
0
50
100
150
200
Time (hr)

Cumulative risk of death plot for wild-type (WT) and TDP-43 mutant (M337V) human iNeurons treated with AMX0114 (0.01-1μM).

Figure 3. Dose-Dependent Decrease in Extracellular NfL Levels Following Treatment with AMX0114

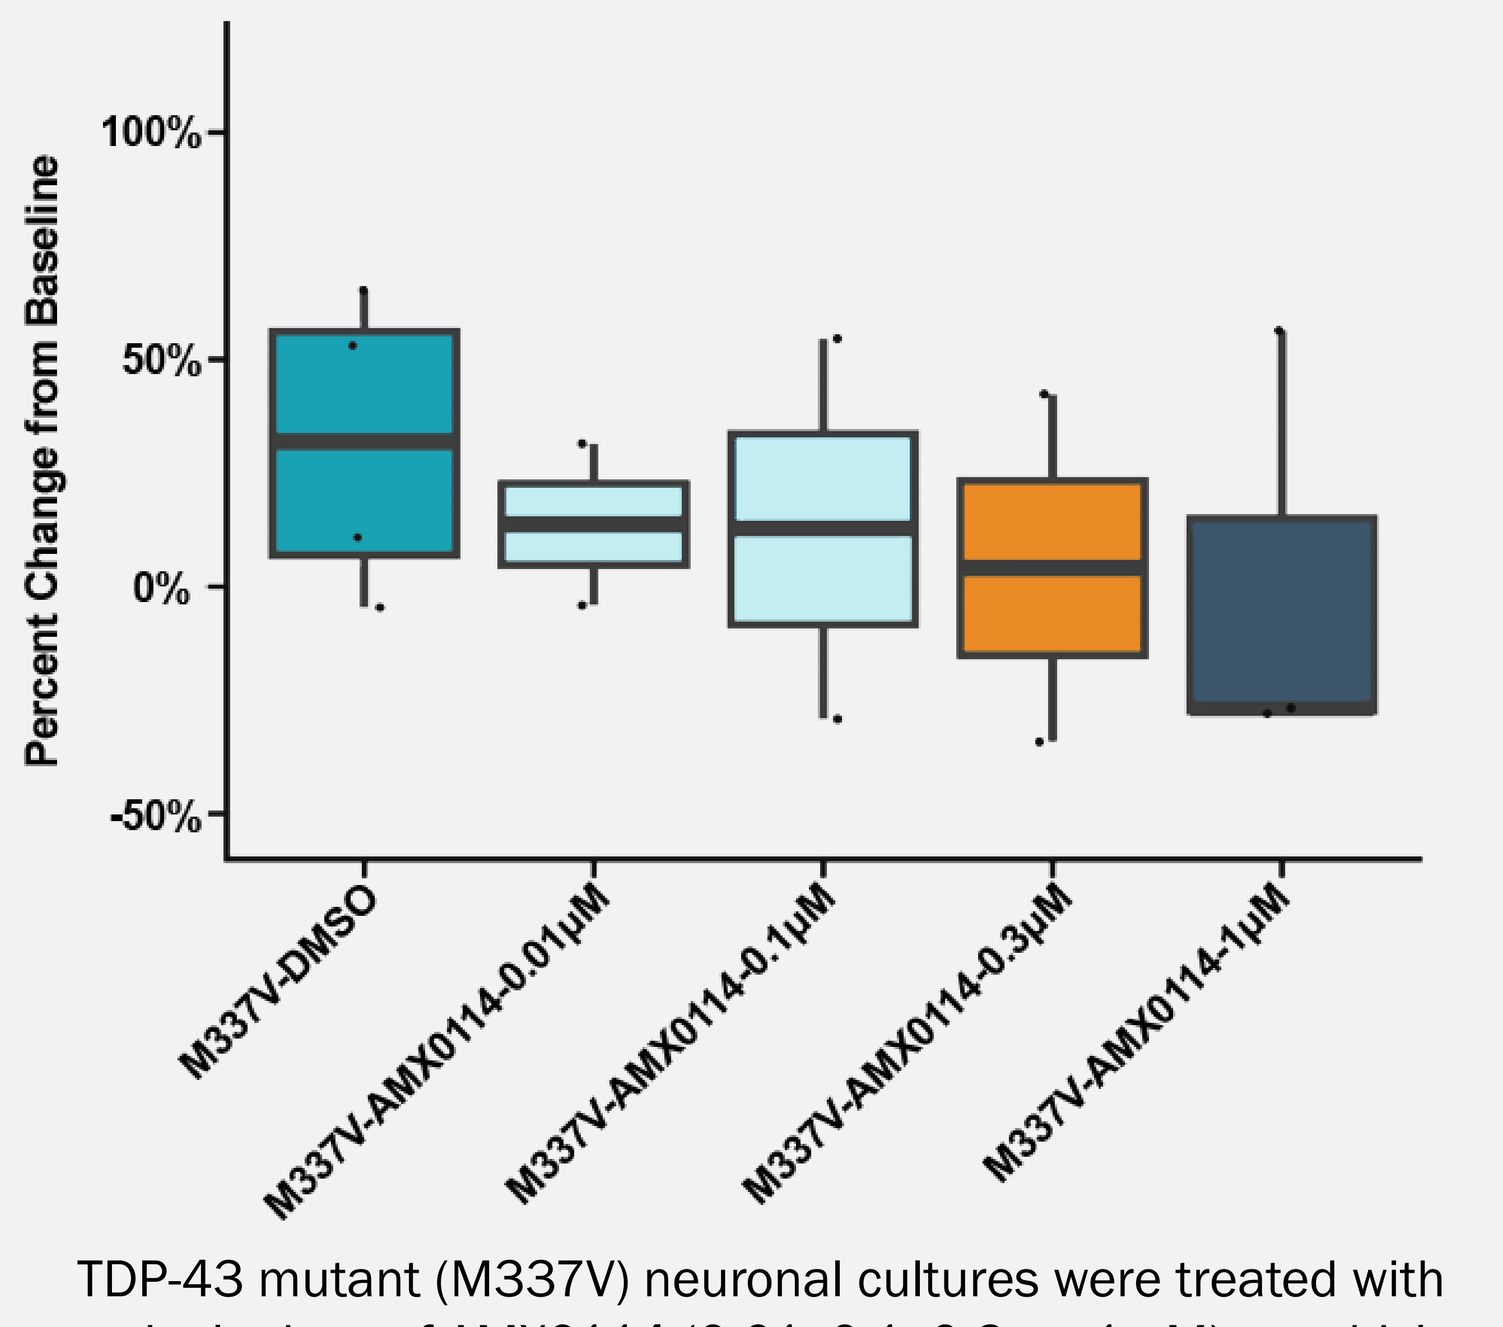
Percent Change from Baseline
100%
50%
0%
-50%
M337V-DMSO
M337V-AMX0114-0.01μM
M337V-AMX0114-0.1μM
M337V-AMX0114-0.3μM
M337V-AMX0114-1μM

TDP-43 mutant (M337V) neuronal cultures were treated with a single dose of AMX0114 (0.01, 0.1, 0.3, or 1 μM) or vehicle (DMSO). Medium was harvested 10 days later and NfL levels were assessed by ELISA. "Baseline" refers to NfL levels in isogenic controls.

In Vitro Efficacy Assessment: Oxidant-Mediated Neuronal Injury

METHOD: Evaluation of cell survival in iPSC-derived motor neurons pre-treated with AMX0114 prior to hydrogen peroxide exposure

- In a model of acute neuronal injury, FujiFilm iCell motor neurons were pre-treated with AMX0114 for 72 hours prior to a time series survival analysis
- After pre-treatment with AMX0114, the cells were exposed to a 95 μM hydrogen peroxide (H_2O_2) dose (at 24h) and 105 μM H_2O_2 dose (at 72h) to cause oxidative stress-induced cell death
- Longitudinal timelapse imaging was used to quantify cell survival

- Treatment of iPSC-derived motor neurons with AMX0114 lowered *CAPN2* mRNA levels by 95.1% and actin-normalized calpain-2 protein levels by 94.4% relative to untreated controls as measured at 120h timepoint
- 96 hours after first exposure to H_2O_2 (120-hour timepoint), 77.2% cell body area remained in the AMX0114-treated neurons, relative to only 12.9% in controls ($p=0.0153$) (**Figure 4**)
- At the latest timepoint tested (192 hours), AMX0114-treated neurons preserved 56% of their initial cell body area, relative to only 11.8% in controls ($p=0.0183$) (**Figure 4**)
- Pre-treatment with AMX0114 achieved potent calpain-2 inhibition and resulted in statistically significant neuroprotection in a model of oxidative stress-induced axonal degeneration (**Figure 4 and 5**)

Figure 4. Full Time Series Survival Analysis

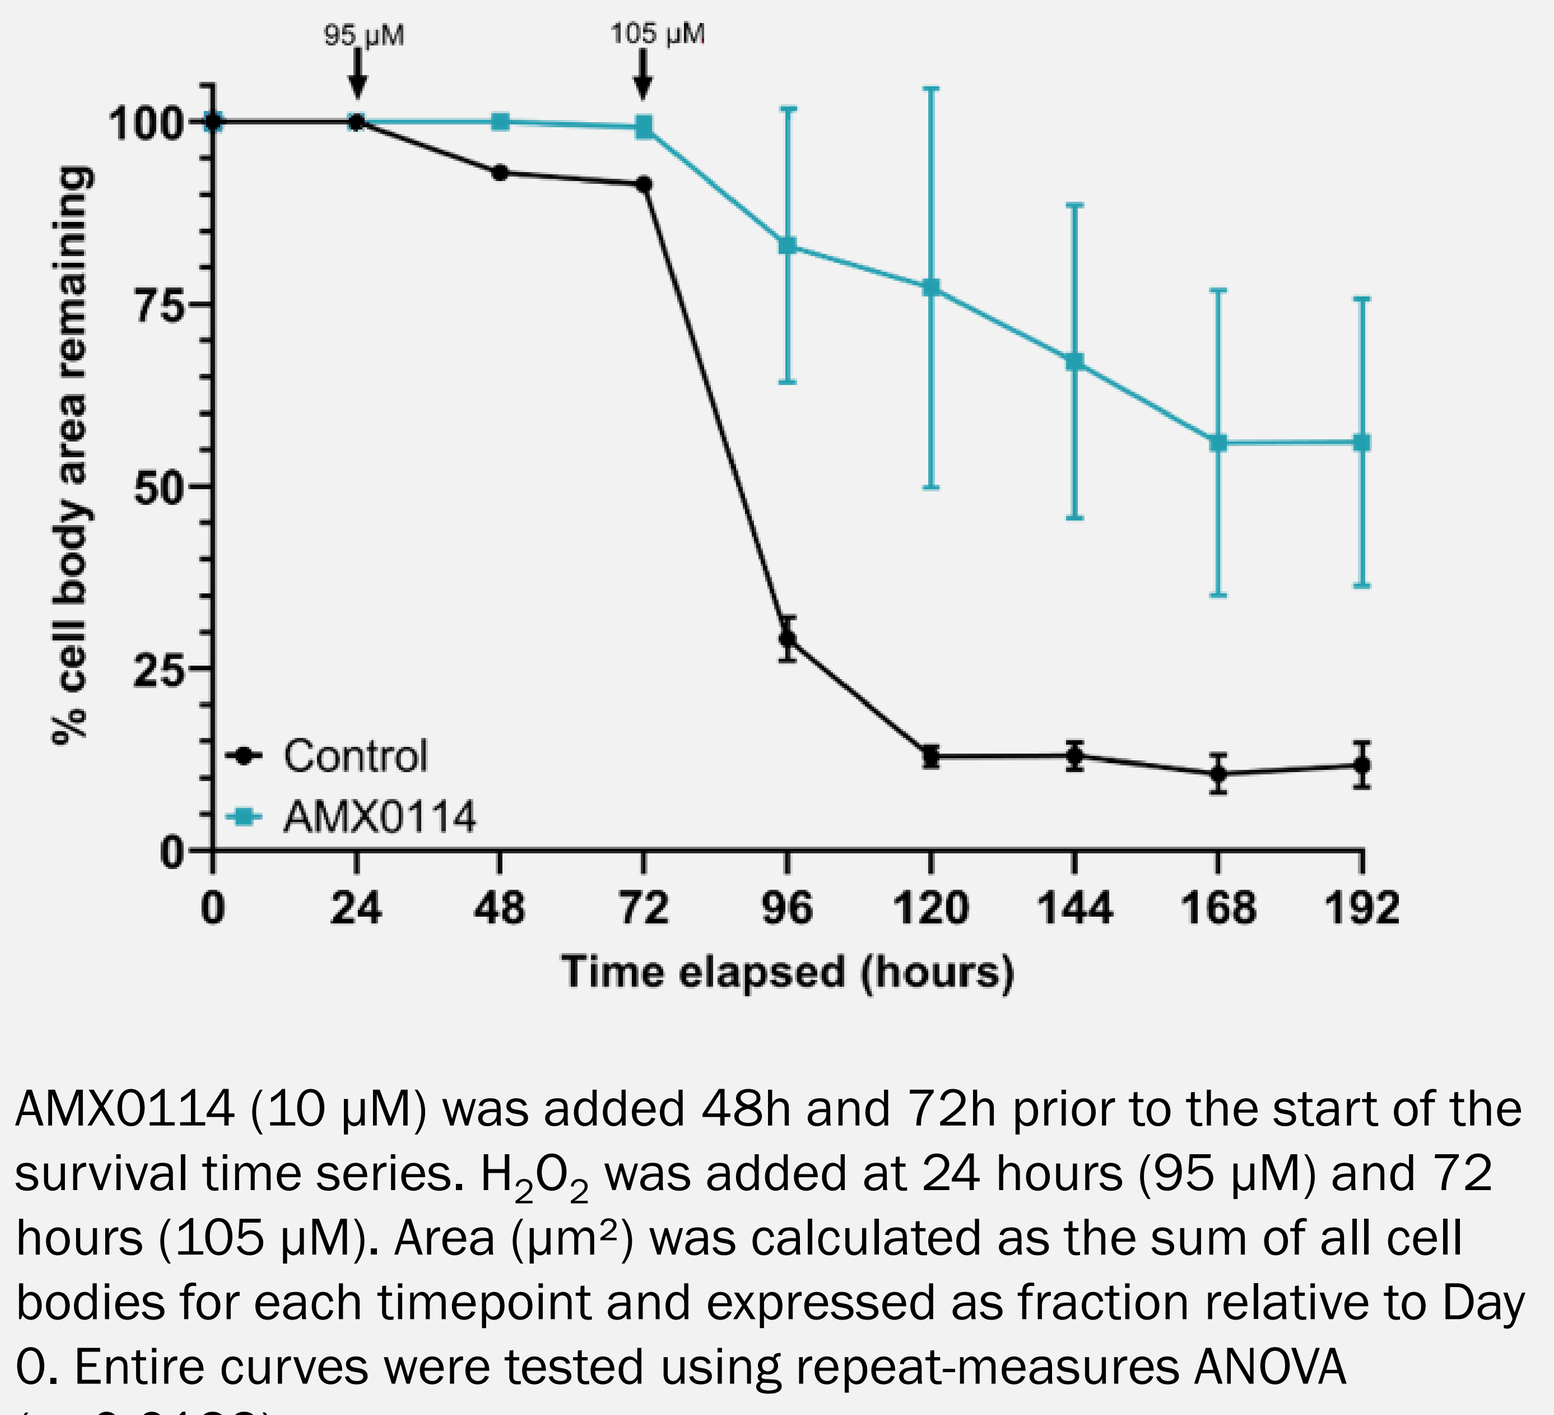
95 μM
105 μM
% cell body area remaining
100
75
50
25
0
Control
AMX0114
0
24
48
72
96
120
144
168
192
Time elapsed (hours)

AMX0114 (10 μM) was added 48h and 72h prior to the start of the survival time series. H_2O_2 was added at 24 hours (95 μM) and 72 hours (105 μM). Area (μm²) was calculated as the sum of all cell bodies for each timepoint and expressed as fraction relative to Day 0. Entire curves were tested using repeat-measures ANOVA ($p=0.0122$).

Figure 5. Representative Images of Motor Neurons Pre- and Post-Exposure to H_2O_2

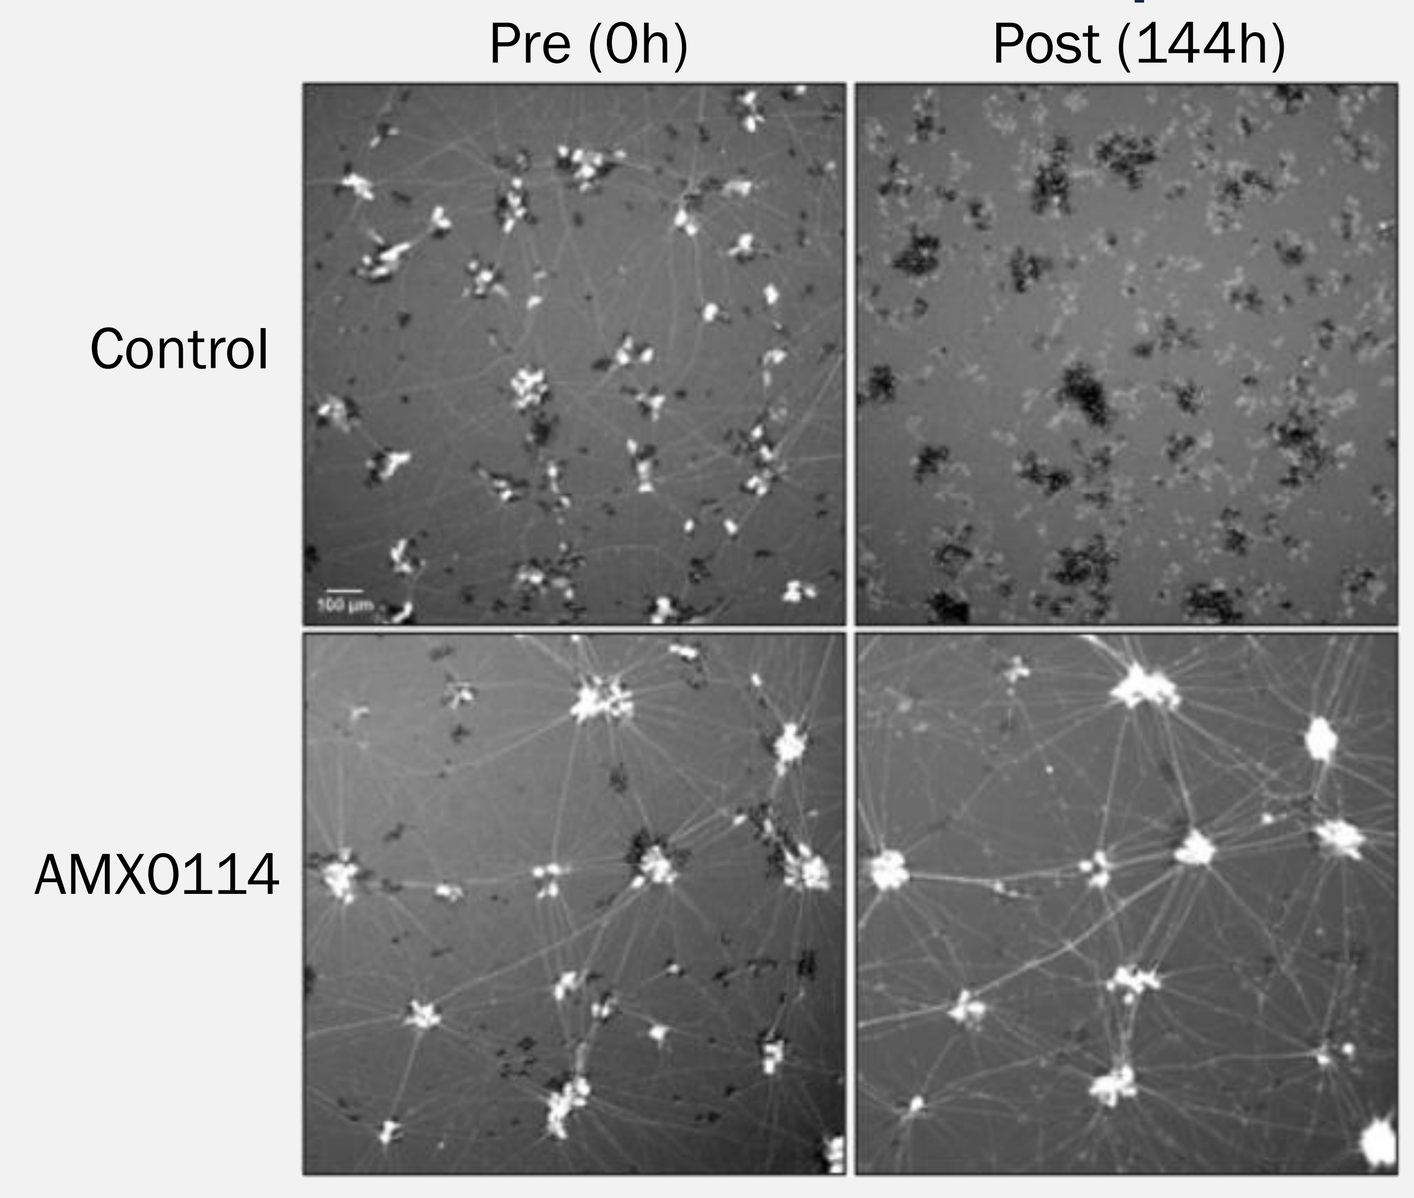
Pre (0h)
Post (144h)
Control
AMX0114

Motor neurons with or without AMX0114 pre-treatment pre-and post H_2O_2 exposure (144-hour timepoint).

CONCLUSIONS

- AMX0114 achieves robust knockdown of *CAPN2* mRNA and calpain-2 protein across multiple disease-relevant cell types and models of axonal degeneration and neuronal death
- This target engagement translates to dose-dependent improvements in neuronal survival and levels of a validated fluid biomarker of neurodegeneration
- Collectively, these data support the hypothesis that reduction of calpain-2 levels is a promising therapeutic strategy in ALS and other neurodegenerative diseases
- AMX0114 has completed IND-enabling studies, and Amylyx expects to initiate a clinical trial studying AMX0114 in ALS in the second half of 2024

Acknowledgements

These studies are sponsored by Amylyx Pharmaceuticals, Inc.

Disclosures

EM is a full-time employee of, and has stock option ownership in Amylyx Pharmaceuticals, Inc. JC and JK are co-CEOs of and own stock in Amylyx Pharmaceuticals, Inc. MB and SB are employees of Michigan Medicine and KM and HK are employees of Weill Cornell Medicine contracted by Amylyx Pharmaceuticals, Inc. to perform the experiments described herein.

References

1. Suzuki N, et al. *Front Neurosci*. 2020;14:194. 2. Ma M. *Neurobiol Dis*. 2013;60:61-79. 3. Asakawa K, et al. *Cell Mol Life Sci*. 2021;78(10):4453-4465. 4. Ueyama H, et al. *J Neurol Sci*. 1998;155(2):163-169. 5. Yamashita T, et al. *Nat Commun*. 2012;3:1307. 6. Rao MV, et al. *J Neurochem*. 2016;137(2):253-265. 7. Wang Y, et al. *Expert Opin Ther Targets*. 2018; 22(1):19-29. 8. Wang JT, et al. J *Cells Bio*. 2012;196(1):7-18.

Presented at TIDES USA 2024; May 14-17, 2023; Boston, MA